Determination of Metformin in Rat Plasma by HILIC-MS/MS Combined with Tecan Automation and Direct Injection

Wei Zhang[1], Futian Han[1], Harry Zhao[1], Zhongping (John) Lin[1], Mike-Qingtao Huang[2], Naidong Weng[2]

[1.] Frontage Laboratories, Inc., 105 Great Valley Parkway, Malvern, PA 19355 [2.] Johnson & Johnson Pharmaceutical Research & Development, LLC, 1000 Route 202 South, Raritan, NJ 08869

Overview

- A validated method for the determination of metformin in rat plasma using HILIC chromatography and tandem mass spectrometry.
- Increased speed and throughput by using Tecan liquid handling system.
- Streamlined the sample extraction process by using direct injection of supernatant after protein precipitation.

Assay Summary

- Simple sample extraction PPT using 96-well plate format
- Automatic liquid handling by Tecan system
- Plasma precipitated with 500 µL of ACN
- Direct organic injection after one-step dilution with ACN
- Calibration curve range: 50 to 50000 ng/mL
- Sample volume: 20 µL
- Method: ESI(+)-HILIC-MS/MS

Introduction

Metformin is a well-know oral antihyperglycaemic used in treatment of Type 2 diabetes. Analysis of metformin is challenging due to the high polarity, small molecular size and the poor retention of metformin on reversed phase column. Hydrophilic Interaction Liquid Chromatography (HILIC) has advantage over reversed phase chromatography in retaining highly polar analytes, and therefore is the method of choice for metformin analysis.
A novel high throughput method was developed and validated for the determination of metformin in rat plasma using hydrophilic interaction liquid chromatography tandem mass spectrometry (HILIC-MS/MS) combined with Tecan automation and direct injection of extracted samples onto silica column in support of preclinical toxicity studies.

Figure 1. Chemical Structures of Metformin and Metformin-d_6

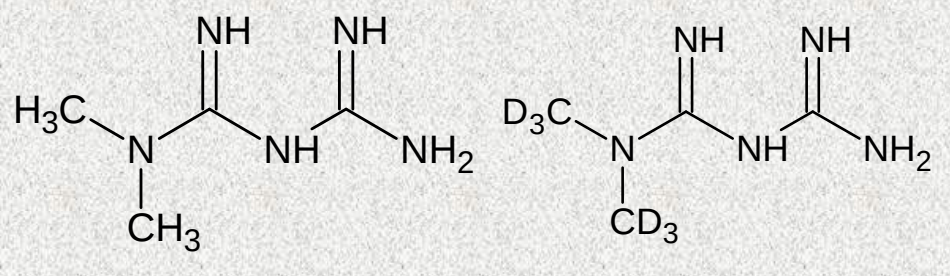

Metformin
MW 129.16

Metformin-d_6 (IS)
MW 135.16

Methods

Chromatographic Conditions

- Column: Luna Silica (2) 100 Å, 50 x 2.0 mm, 3µ
- Mobile Phase: 2 mM NH_4COOH & 0.1 % HCOOH in ACN/H_2O (80/20, v/v)
- Flow Rate: 0.4 mL/min
- Inj. Volume: 5 µL
- Needle Wash: 2 mM NH_4COOH & 0.1 %; HCOOH in MeOH/H_2O (50/50, v/v)
- Run Time: 3 minutes

MS/MS Detection

- Mass Spectrometer: Sciex API 4000
- Ionization: Positive Ion Electrospray
- MRM Transitions: Metformin: m/z 130.2 → 70.9
 IS: m/z 136.1 → 76.9

Results

Linearity

- Calibration curve range: 50 to 50000 ng/mL
- Correlation coefficient: r ≥ 0.9993

Sensitivity, Specificity and Recovery

- **LLOQ:**
 50 ng/mL, n=6, CV% 5.0 and %Nominal 98.3%
- **Specificity:**
 Six lots blank human plasma were screened and were free of interference.
- **Matrix Factor:**
 IS-normalized MF: 1.03±0.02 (n=6) indicating no significant matrix effect for the assay.
- **Recovery:** 91.0%

Figure 2. Typical chromatogram of LLOQ sample

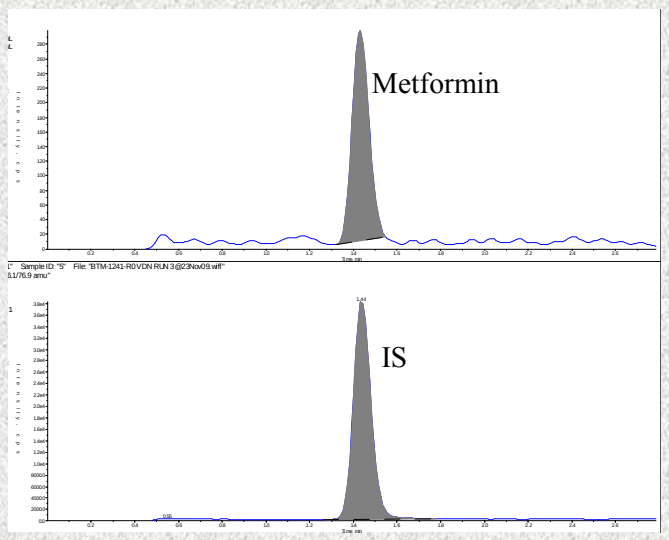
Metformin
IS

Precision and Accuracy

Calibration standards:
%CV < 2.3% and Accuracy: > 97.9 (Table 1)
Intra-run QC results:
%CV < 3.8% and Accuracy: > 98.9 (Table 2)
Inter-run QC results:
%CV < 3.9% and Accuracy: > 99.0 (Table 2)

Table 1. Precision and Accuracy of Calibration Standards

	Metformin Concentration, ng/mL							
N=5	50	100	500	1000	5000	10000	40000	50000
Mean	49.8	101	498	1026	5027	10011	39180	49404
SD	0.5	2.3	6.5	13.4	53.6	85.1	355	1115
%CV	1.1	2.3	1.3	1.3	1.1	0.9	0.9	2.3
% Nominal	99.5	100.8	99.7	102.6	100.5	100.1	97.9	98.8

Table 2. Precision and Accuracy of Validation QC Samples

Run ID	N=6	Metformin Concentration, ng/mL		
		150	4000	39000
Run1	Mean	150	4092	38552
	SD	3.3	53.7	613
	%CV	2.2	1.3	1.6
	%Nominal	99.8	102.3	98.9
Run2	Mean	158	4086	38592
	SD	6.1	19.2	203
	%CV	3.8	0.5	0.5
	%Nominal	106	102.1	99.0
Run3	Mean	149	4133	39427
	SD	4.9	64.1	630
	%CV	3.3	1.6	1.6
	%Nominal	99.4	103.3	101.1
Overall Results	Mean	152	4072	38614
	SD	6.0	73.3	786
	%CV	3.9	1.8	2.0
	%Nominal	101.5	101.8	99.0

Stabilities:

- Bench-top (RT) stability: at least 6 hours.
- Freeze/thaw stability: 3-cycles.
- Processed sample stability: at least 66 hours.
- Long-term stability (-70°C) in human plasma: at least 147 days.
- Whole blood stability (at room temperature and at 0-4 °C): at least 2 hours.

Dilution Integrity

A 20-fold dilution with blank plasma was performed.

Ruggedness

- A complete validation was performed using API 4000
- Results obtained using API 4000: $r^2 \geq 0.9993$; QC results: %CV ≤ 6.1%; Accuracy: 91.4 to 106.0%
- Supported several toxicity studies with excellent QC performance (Table 3)

Table 3. Precision and Accuracy of QC Sample from Several Toxicity Studies

	Metformin Concentration, ng/mL		
	150	4000	39000
Mean	157	4120	37902
SD	7.7	126.5	2516
%CV	4.9	3.1	6.6
%Nominal	104.9	103.0	97.2

Ruggedness was further proved by ISR Evaluation

- Incurred Sample Stability (ISR) has been evaluated using a combined 22 samples from two toxicity studies with 100% of the results met the following criterion.
- Acceptance Criterion: % Diff of at least 2/3 of all of the ISR samples should be within ± 20%.
- The %Diff of the result of the ISR analysis was calculated as follows:

%Diff =(Repeat - Original)/(Mean of Original and Repeat) x 100

Conclusions

A simple automatic, high throughput, HILIC-MS/MS method for determination of metformin in rat plasma has been developed and validated.
The method was fast, robust, and reliable. The validated high throughput method has been successfully used in support of several toxicity studies.